Subgroup analysis of patients with no prior chemotherapy in EMERALD: A phase 3 trial evaluating elacestrant, an oral selective estrogen receptor degrader (SERD), vs investigator's choice of endocrine monotherapy for ER+/HER2- advanced/metastatic breast cancer (mBC)

Kaklamani V,[1] Bardia A,[2] Aftimos P,[3] Cortes J,[4] Lu J,[5] Neven P,[6] Streich G,[7] Montero AJ,[8] Forget F,[9] Mouret-Reynier MA,[10] Sohn JH,[11] Taylor D,[12] Harnden KK,[13] Khong H,[14] Kocsis J,[15] Dalenc F,[16] Dillon P,[17] Tonini G,[18] Grzegorzewski KJ,[19] Bidard FC[20]

[1]University of Texas Health Sciences Center, Houston, TX; [2]Massachusetts General Hospital Cancer Center, Harvard Medical School, Boston, MA, USA; [3]Institut Jules Bordet – Université Libre de Bruxelles, Brussels, Belgium; [4]International Breast Cancer Center (IBCC), Quiron Group, Barcelona, Spain; [5]University Of Southern California/Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [6]Universitaire Ziekenhuizen (UZ) – Leuven Cancer Institute, Leuven, Belgium; [7]Centro Médico Austral, Buenos Aires, Argentina; [8]University Hospitals Seidman Cancer Center – Case Western Reserve University, Cleveland, OH, USA; [9]Centre Hospitalier de l'Ardenne – Site de Libramont, Libramont-Chevigny, Belgium; [10]Centre Jean Perrin, Clermont-Ferrand, France; [11]Yonsei Cancer Center, Yonsei University Health System – Medical Oncology, Seoul, Republic of Korea; [12]Université catholique de Louvain, CHU UCL Namur – Site Sainte-Elisabeth, Namur, Belgium; [13]Inova Schar Cancer Institute, Fairfax, Virginia; [14]Moffit Cancer Center & Research Institute, Tampa, FL, USA; [15]Bács-Kiskun Megyei Kórház, Kecskemét, Hungary; [16]Institut Claudius Regaud, IUCT-Oncopole, Toulouse, France; [17]University of Virginia Cancer Center, Charlottesville, VA, USA; [18]Menarini Group, Florence, Italy; [19]Stemline Therapeutics/Menarini Group, New York, NY, USA; [20]Institut Curie, Paris and Saint Cloud, France

Presented at ASCO® 2022

Abstract: 1100

Poster Selected for the 2022 GRASP Advocate Choice Award

EMERALD

BACKGROUND

- Endocrine therapy, with aromatase inhibitor (AI) or fulvestrant, plus cyclin-dependent kinase 4/6 inhibitor (CDK4/6i) is the recommended first-line treatment of estrogen receptor-positive (ER+)/HER2- mBC.[1-3]
- Subsequent disease progression is associated with endocrine resistance, including the development of *ESR1* mutations (*mESR1*).[4]
- Treatment guidelines recommend use of sequential endocrine therapy before chemotherapy, in the absence of visceral crisis or until all endocrine therapy (ET) options have been exhausted.[1-3, 5]
- Standard single-agent endocrine therapy (eg, fulvestrant) in patients who have received prior CDK4/6i or mTOR inhibitor is associated with poor median progression-free survival (~2 months),[6-9] highlighting a major unmet need for patients with ER+/HER2- mBC.
- Elacestrant (RAD1901) is an oral SERD that blocks ER and inhibits estradiol-dependent gene transcription induction and cell proliferation in ER+ BC cell lines with higher efficacy than fulvestrant.[10]
- In a phase 3 study of elacestrant in postmenopausal women with ER+/HER2- mBC (EMERALD), elacestrant significantly reduced the risk of disease progression or death by 30% in all patients and by 45% in patients with *ESR1* mutation (Figure 1a & b).[11]
- In this analysis, we compared PFS between elacestrant and SOC in patients without prior chemotherapy in the metastatic setting.

Figure 1a: PFS in all patients (ITT) (N=477)

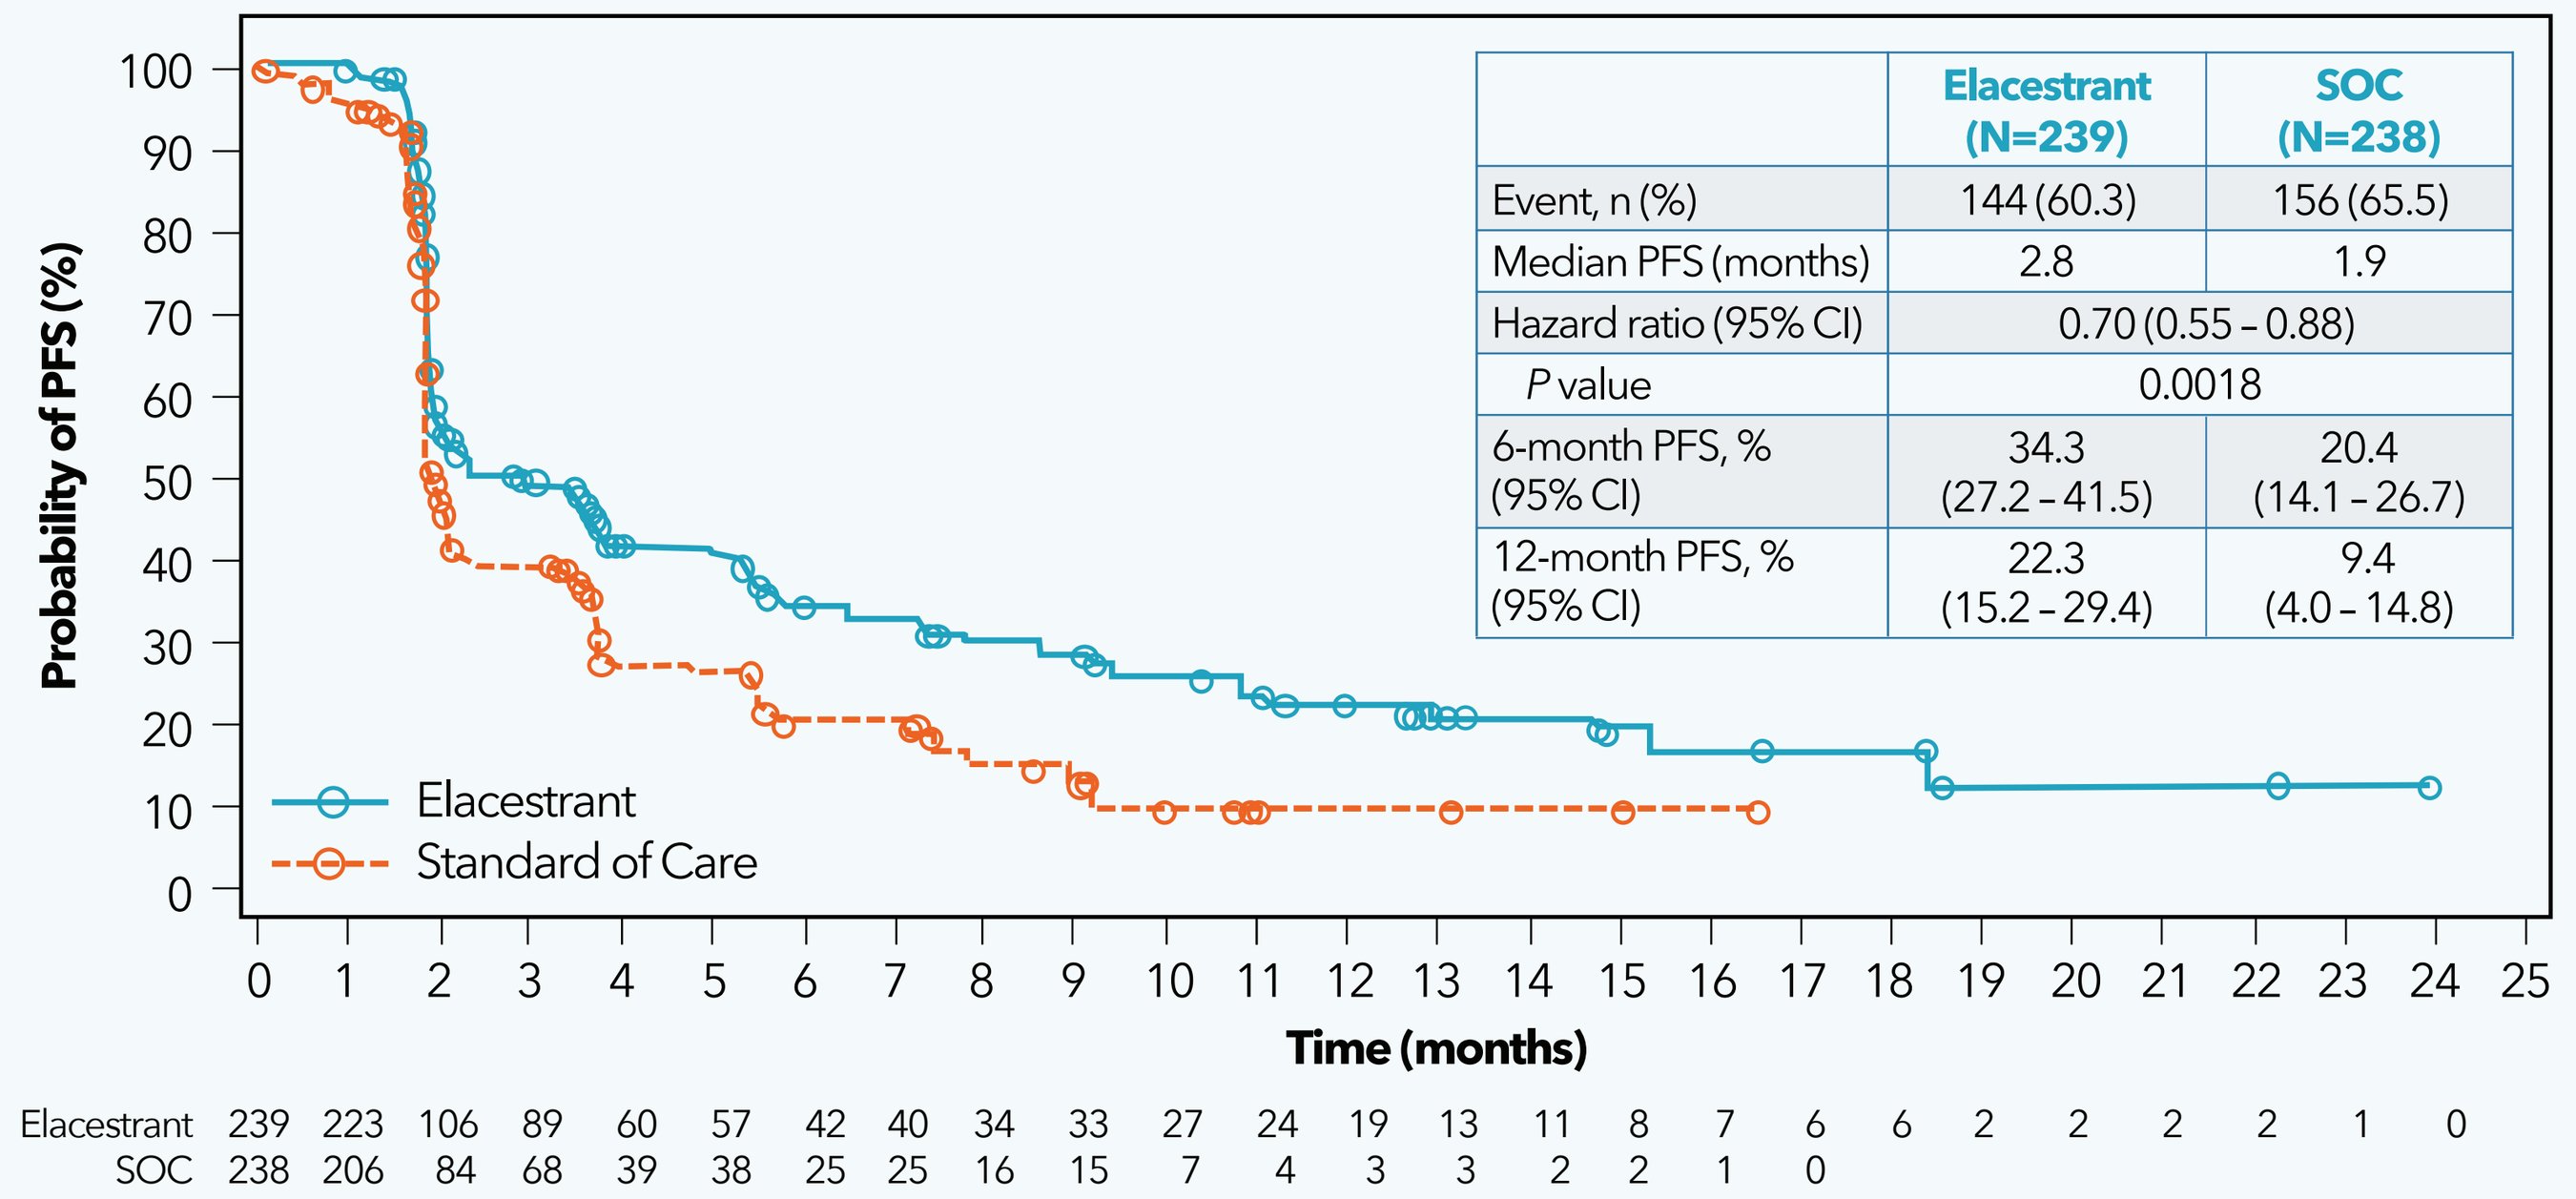

Probability of PFS (%)
Time (months)
Elacestrant
Standard of Care
Elacestrant 239 223 106 89 60 57 42 40 34 33 27 24 19 13 11 8 7 6 6 2 2 2 2 1 0
SOC 238 206 84 68 39 38 25 25 16 15 7 4 3 3 2 2 1 0

	Elacestrant (N=239)	SOC (N=238)
Event, n (%)	144 (60.3)	156 (65.5)
Median PFS (months)	2.8	1.9
Hazard ratio (95% CI)	0.70 (0.55 – 0.88)	
P value	0.0018	
6-month PFS, % (95% CI)	34.3 (27.2 – 41.5)	20.4 (14.1 – 26.7)
12-month PFS, % (95% CI)	22.3 (15.2 – 29.4)	9.4 (4.0 – 14.8)

Figure 1b: PFS in all patients with detectable m*ESR1* (N=228)

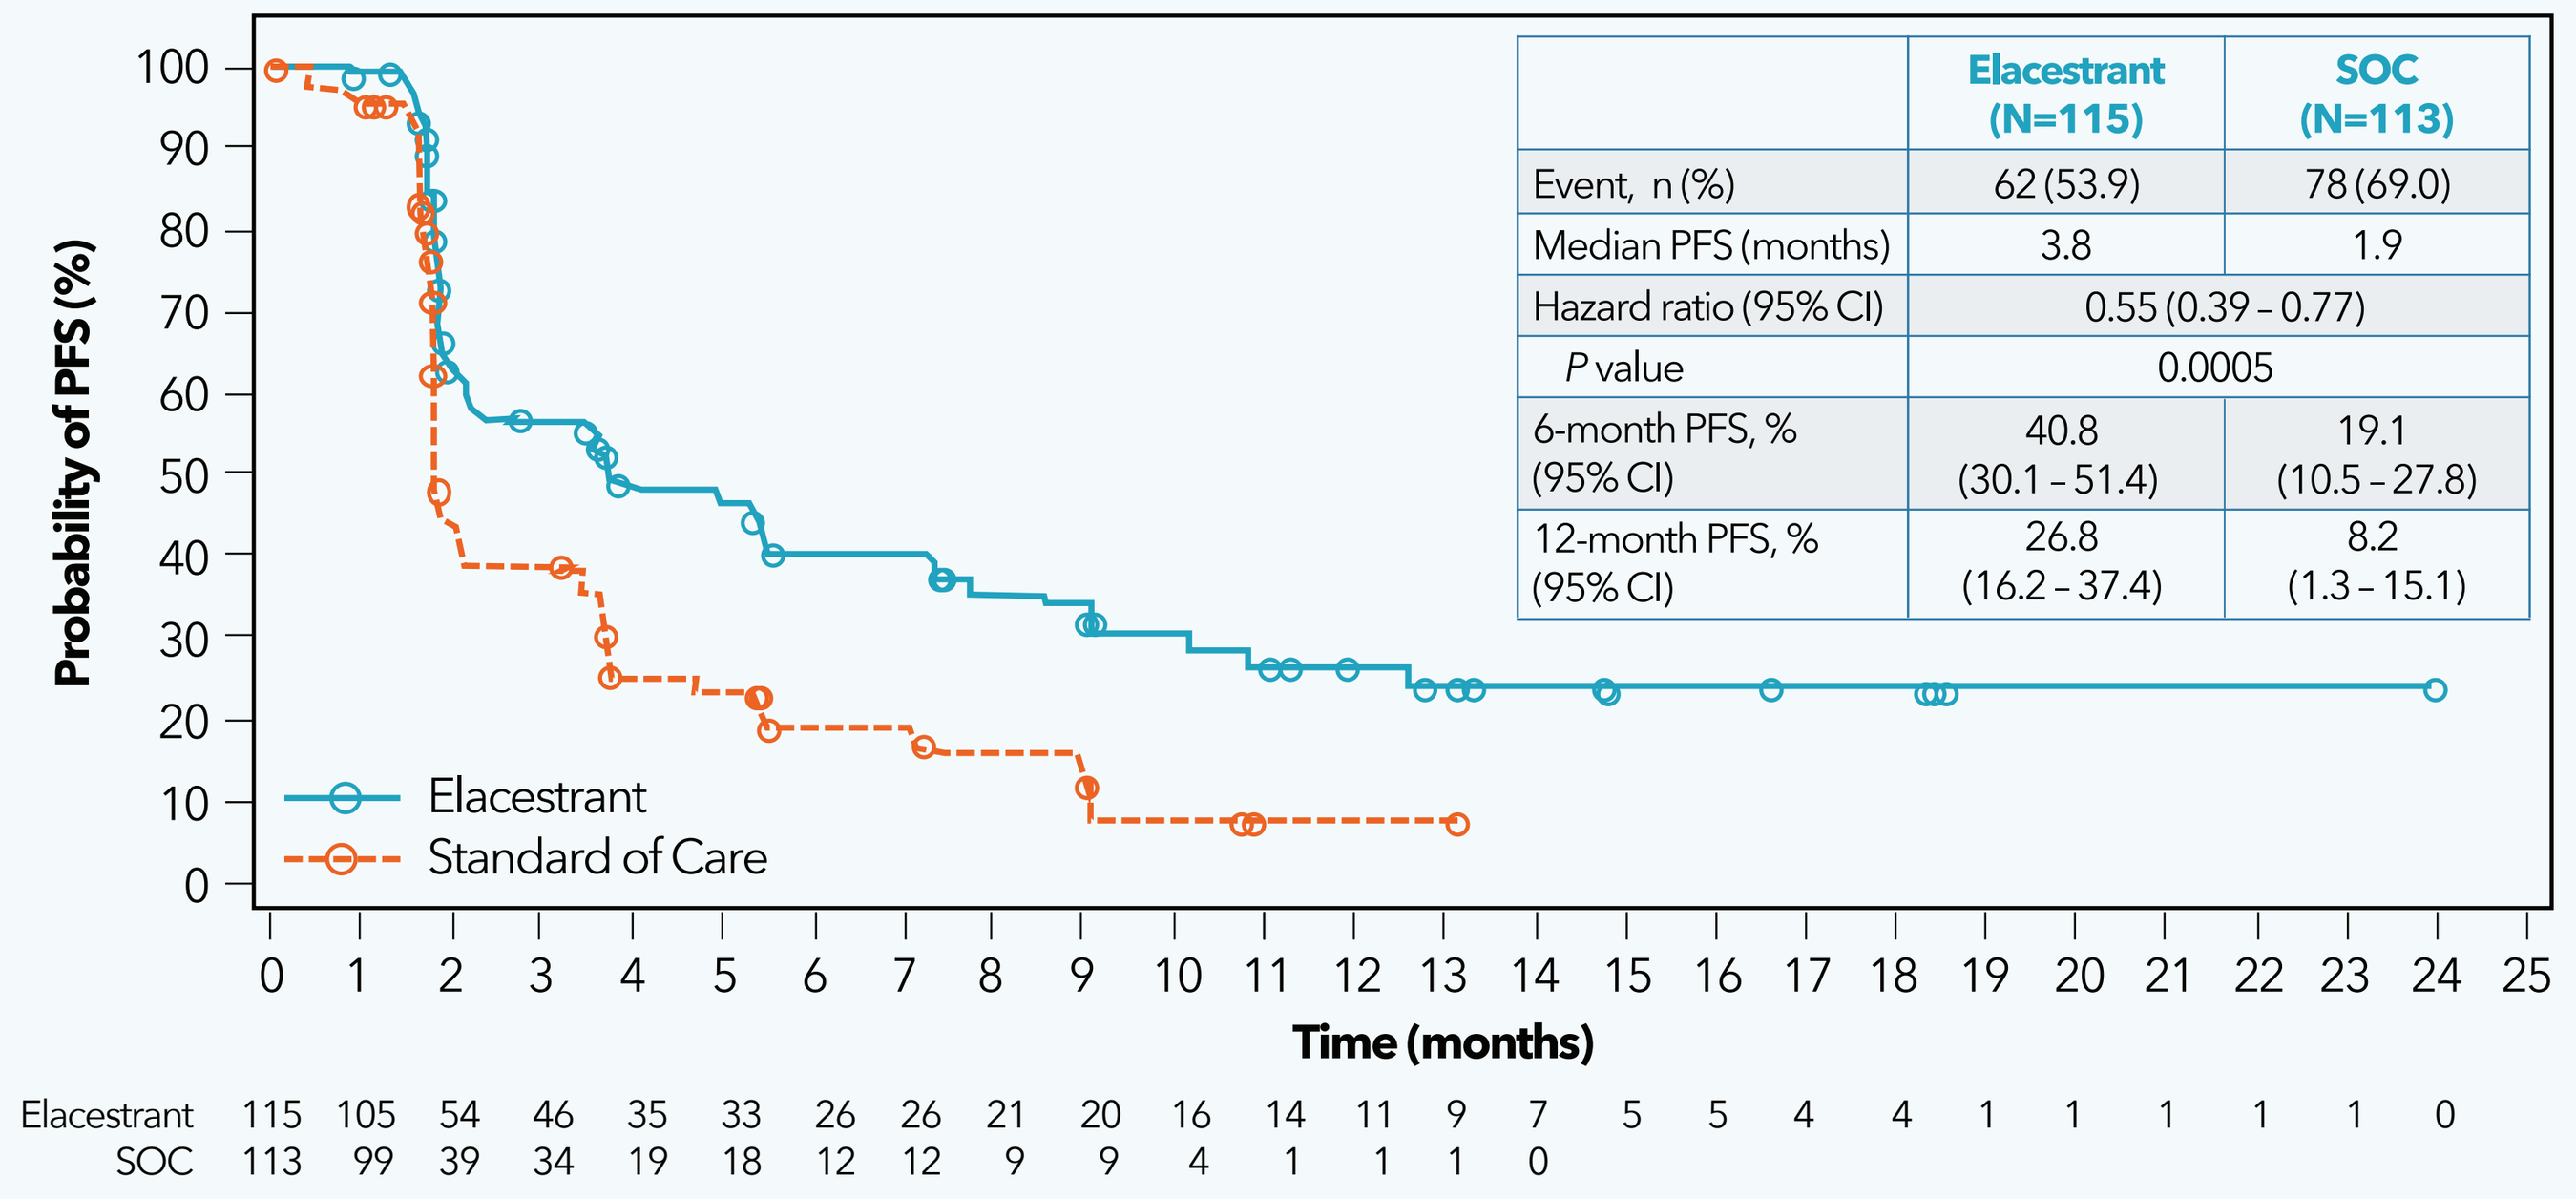

Probability of PFS (%)
Time (months)
Elacestrant
Standard of Care
Elacestrant 115 105 54 46 35 33 26 26 21 20 16 14 11 9 7 5 5 4 4 1 1 1 1 1 0
SOC 113 99 39 34 19 18 12 12 9 9 4 1 1 1 0

	Elacestrant (N=115)	SOC (N=113)
Event, n (%)	62 (53.9)	78 (69.0)
Median PFS (months)	3.8	1.9
Hazard ratio (95% CI)	0.55 (0.39 – 0.77)	
P value	0.0005	
6-month PFS, % (95% CI)	40.8 (30.1 – 51.4)	19.1 (10.5 – 27.8)
12-month PFS, % (95% CI)	26.8 (16.2 – 37.4)	8.2 (1.3 – 15.1)

EMERALD STUDY DESIGN[12]

Inclusion Criteria
- Men and postmenopausal women with advanced/ metastatic breast cancer
- ER-positive,[a] HER2-negative
- Progressed or relapsed on or after 1 or 2 lines of endocrine therapy for advanced disease, one of which was given in combination with a CDK4/6i
- ≤1 line of chemotherapy for advanced disease
- ECOG PS 0 or 1

N=477[b]

R 1:1

Elacestrant 400 mg daily[c]

Investigator's choice (SOC): Fulvestrant, Anastrozole, Letrozole, Exemestane

PD or withdrawal criterion → Follow-up

Primary Endpoints:[d]
- PFS in all patients
- PFS in *mESR1*

Key Secondary Endpoints:
- OS in all patients
- OS in *mESR1*

Other Secondary Endpoints:
- PFS and OS in patients without *mESR1*[f]
- PFS by investigator
- ORR
- DOR
- CBR
- Safety and tolerability

Stratification Factors:
- *ESR1*-mutation status[e]
- Prior treatment with fulvestrant
- Presence of visceral metastases

SOC guidance recommended use of a different ET than previously received (ie, fulvestrant recommended for patients who had not previously received fulvestrant, and selection of AI was based on prior AI therapy)

[a]Documentation of ER+ tumor with ≥ 1% staining by immunohistochemistry (local laboratory); [b]Recruitment from February 2019 to October 2020; [c]Protocol-defined dose reductions permitted; [d]Blinded Independent Central Review; [e]*ESR1*-mutation status was determined by cell-free circulating DNA analysis using the Guardant360® CDx (Guardant Health, Redwood City, CA); [f]Restaging CT scans every 8 weeks.
CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; ER, estrogen receptor; HER2, human epidermal growth factor receptor 2; *mESR1*, *ESR1*-mutant breast cancer population; PFS, progression-free survival; R, randomized; SOC, standard of care.

RESULTS

Baseline demographic and disease characteristics

Among the 477 patients enrolled in the trial, 77.8% (n=371) had not received prior chemo for mBC

Parameter	Elacestrant All (N=191)	Elacestrant *mESR1* (N=89)	SOC All (N=180)	SOC *mESR1* (N=81)
Median age, years (range)	64 (28-89)	64 (28-89)	64 (35-83)	63 (35-83)
Gender, n %				
Female	185 (96.9)	89 (100)	180 (100)	81 (100)
Male	6 (3.1)	0	0	0
ECOG PS, n (%)				
0	111 (58.1)	48 (53.9)	100 (55.6)	44 (54.3)
1	80 (41.9)	41 (46.1)	180 (44.4)	37 (45.7)
Visceral metastasis*, n (%)	127 (66.5)	62 (69.7)	125 (69.4)	61 (75.3)
Bone-only disease, n (%)	32 (16.8)	10 (11.2)	25 (13.9)	10 (12.3)
Prior adjuvant therapy, n (%)	129 (67.5)	50 (56.2)	114 (63.3)	51 (63.0)
Prior CDK4/6 inhibitor, n (%)	191 (100)	89 (100)	180 (100)	81 (100)
Number of prior lines of endocrine therapy,** n (%)				
1	103 (53.9)	56 (62.9)	115 (63.9)	56 (69.1)
2	88 (46.1)	33 (37.1)	65 (36.1)	25 (30.9)
Number of prior lines of chemotherapy,** n (%)				
0	191 (100)	89 (100)	180 (100)	81 (100)

*Includes lung, liver, brain, pleural, and peritoneal involvement
**In the advanced/metastatic setting

PFS: elacestrant vs SOC in all patients without prior chemotherapy (N=371)

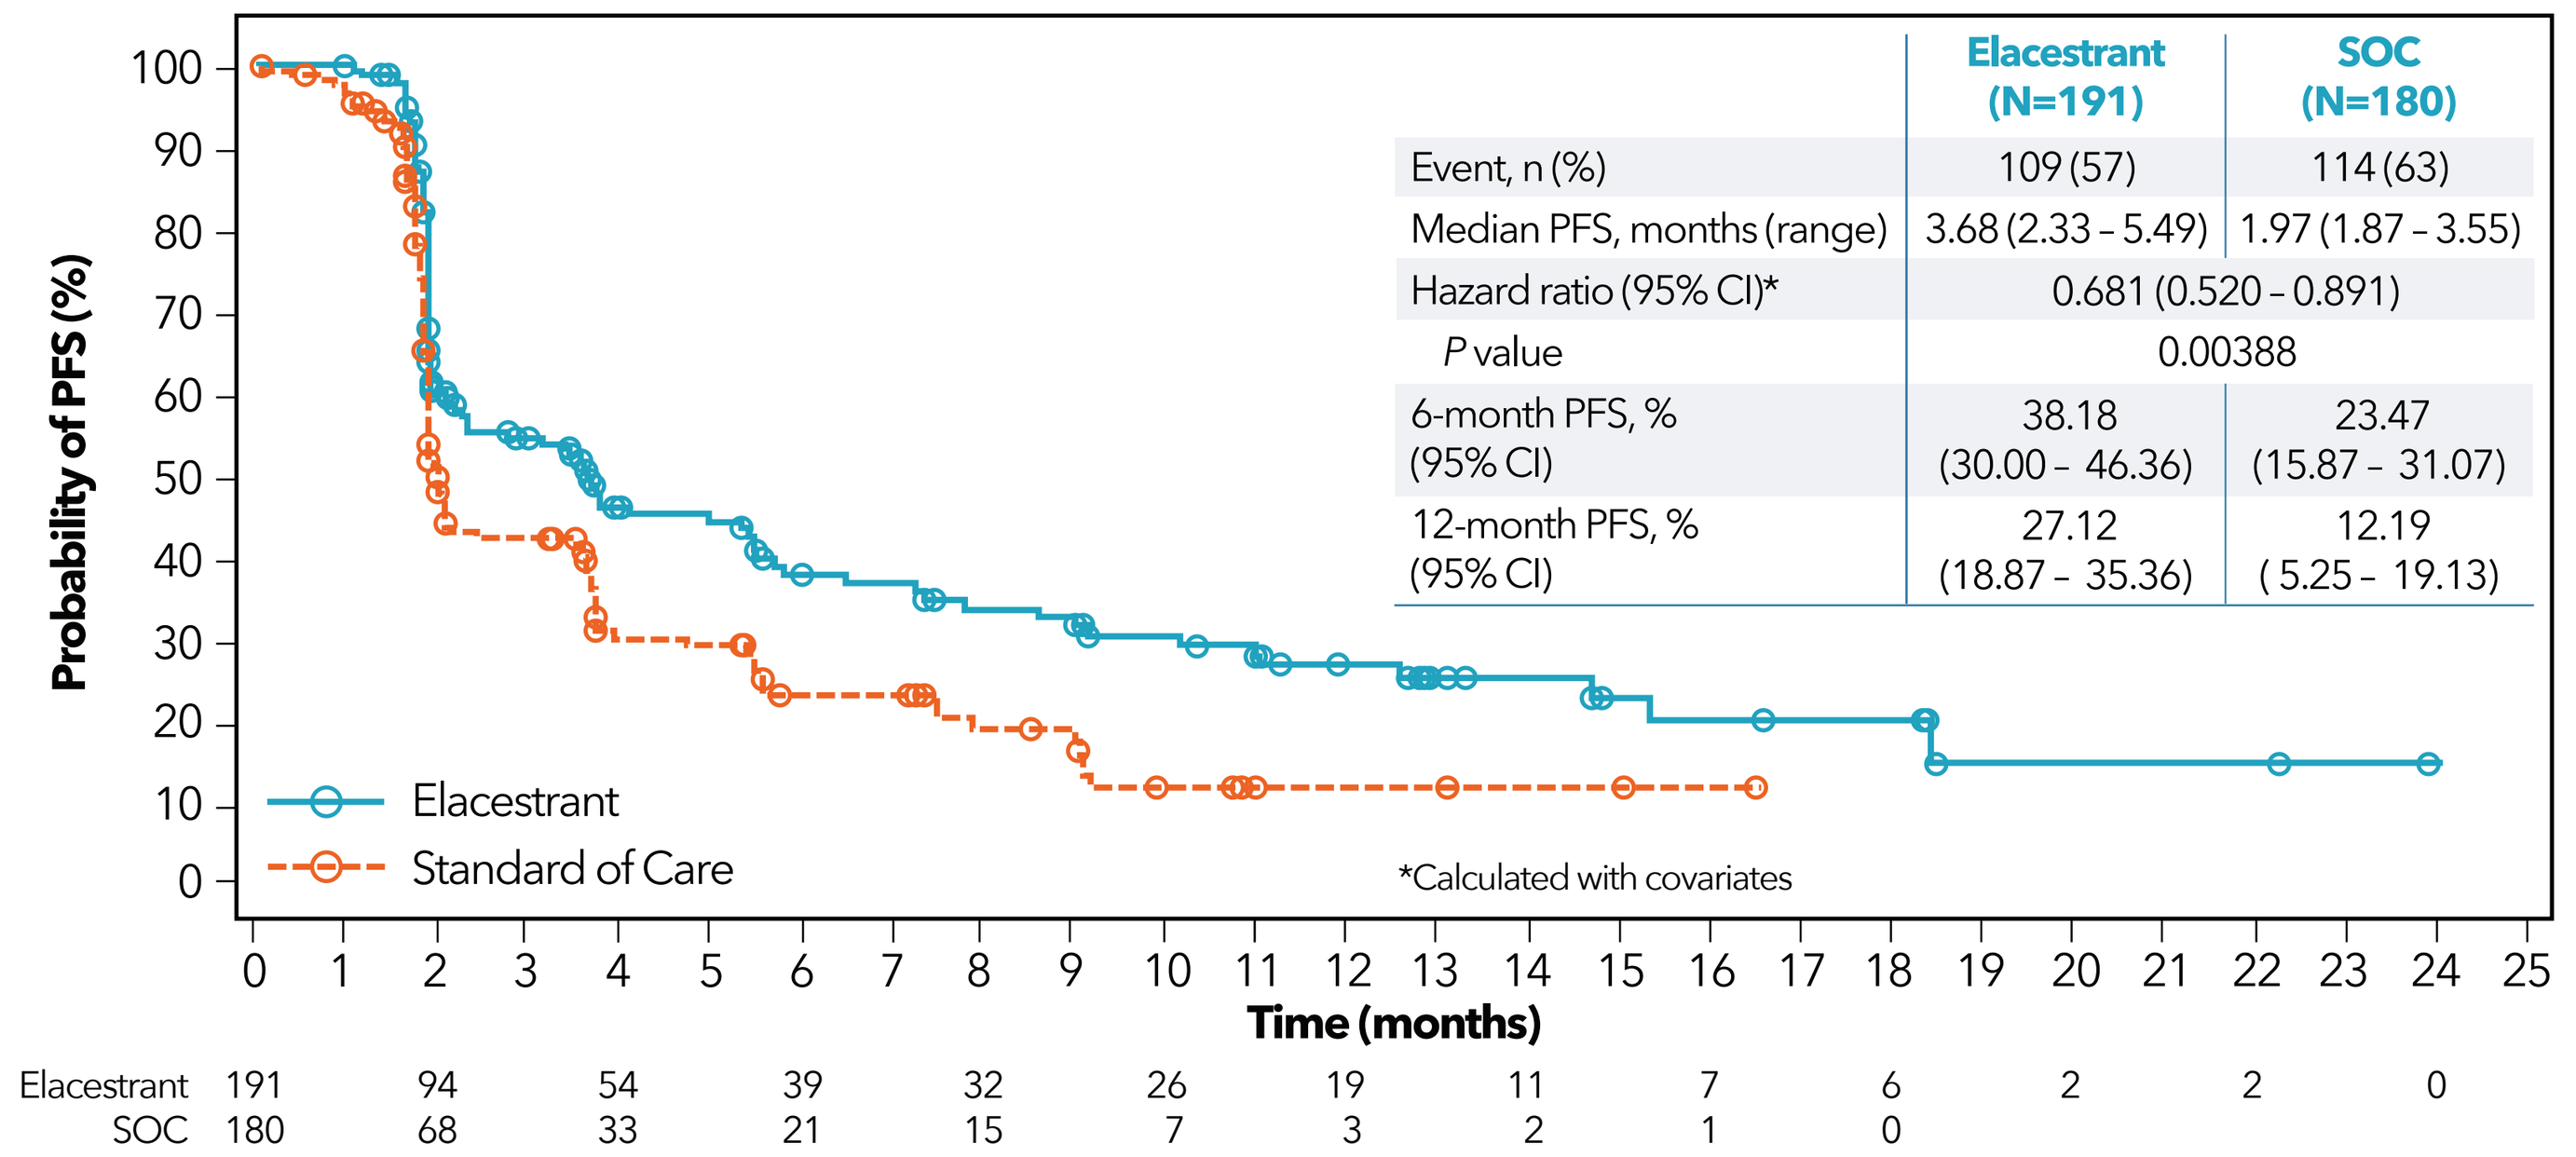

Probability of PFS (%)
Time (months)
Elacestrant
Standard of Care
Elacestrant 191 94 54 39 32 26 19 11 7 6 2 2 0
SOC 180 68 33 21 15 7 3 2 1 0

	Elacestrant (N=191)	SOC (N=180)
Event, n (%)	109 (57)	114 (63)
Median PFS, months (range)	3.68 (2.33 – 5.49)	1.97 (1.87 – 3.55)
Hazard ratio (95% CI)*	0.681 (0.520 – 0.891)	
P value	0.00388	
6-month PFS, % (95% CI)	38.18 (30.00 – 46.36)	23.47 (15.87 – 31.07)
12-month PFS, % (95% CI)	27.12 (18.87 – 35.36)	12.19 (5.25 – 19.13)

*Calculated with covariates

Among patients with ER+/HER2– mBC without prior chemotherapy, elacestrant significantly prolonged PFS compared to SOC

PFS: elacestrant vs SOC in patients with *mESR1* without prior chemotherapy (N=170)

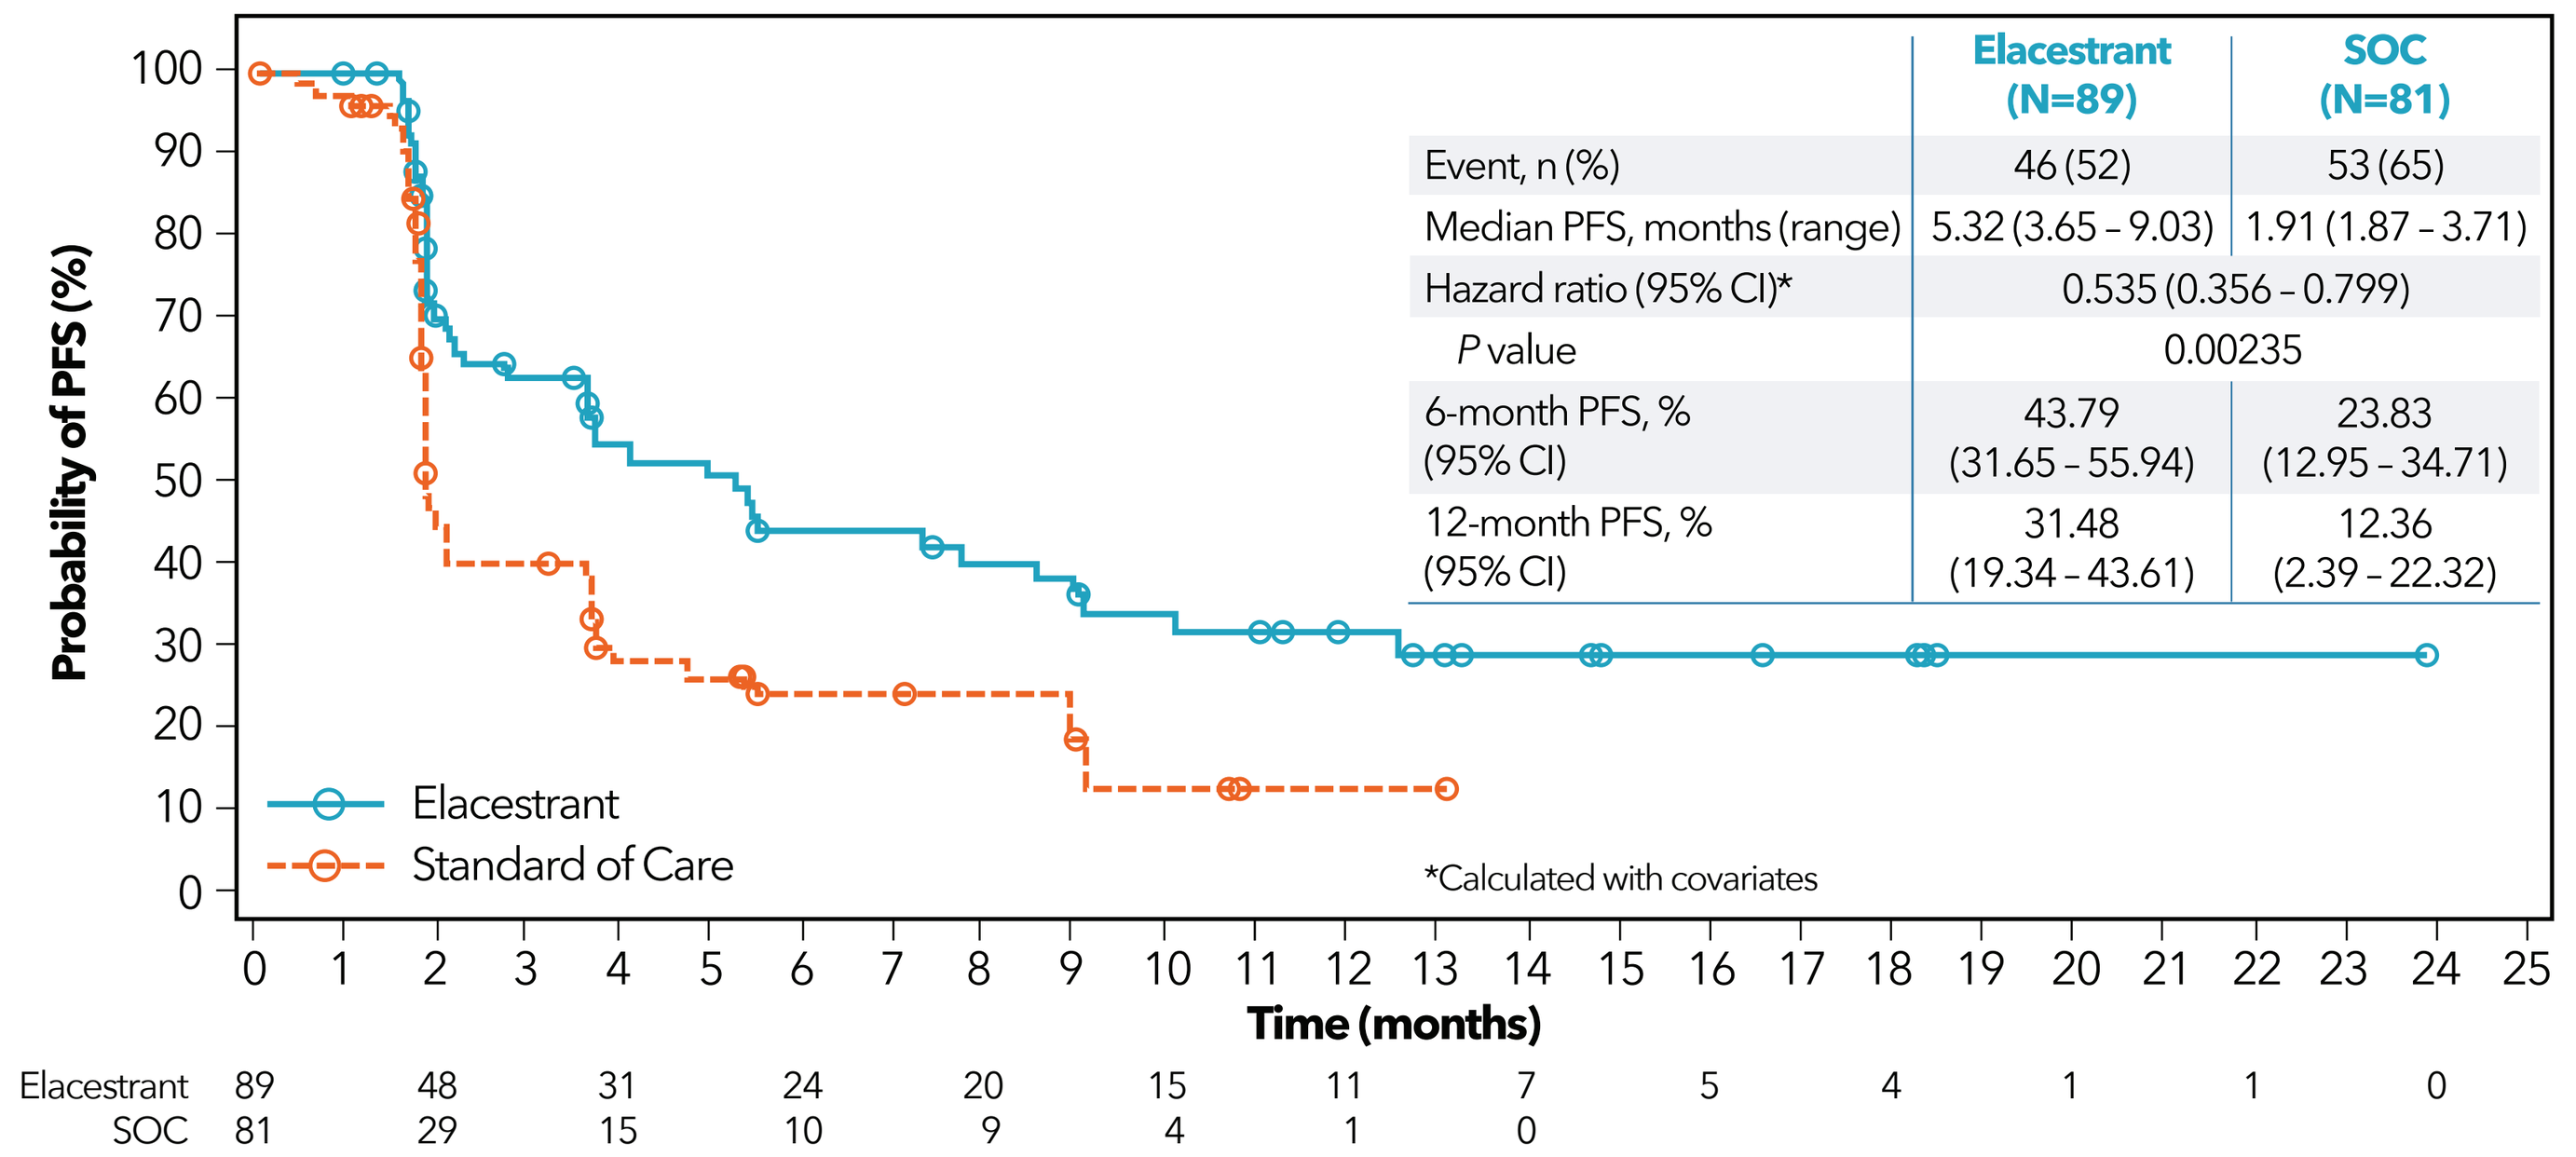

Probability of PFS (%)
Time (months)
Elacestrant
Standard of Care
Elacestrant 89 48 31 24 20 15 11 7 5 4 1 1 0
SOC 81 29 15 10 9 4 1 0

	Elacestrant (N=89)	SOC (N=81)
Event, n (%)	46 (52)	53 (65)
Median PFS, months (range)	5.32 (3.65 – 9.03)	1.91 (1.87 – 3.71)
Hazard ratio (95% CI)*	0.535 (0.356 – 0.799)	
P value	0.00235	
6-month PFS, % (95% CI)	43.79 (31.65 – 55.94)	23.83 (12.95 – 34.71)
12-month PFS, % (95% CI)	31.48 (19.34 – 43.61)	12.36 (2.39 – 22.32)

*Calculated with covariates

PFS: elacestrant vs fulvestrant in all patients without prior chemotherapy (N=323)

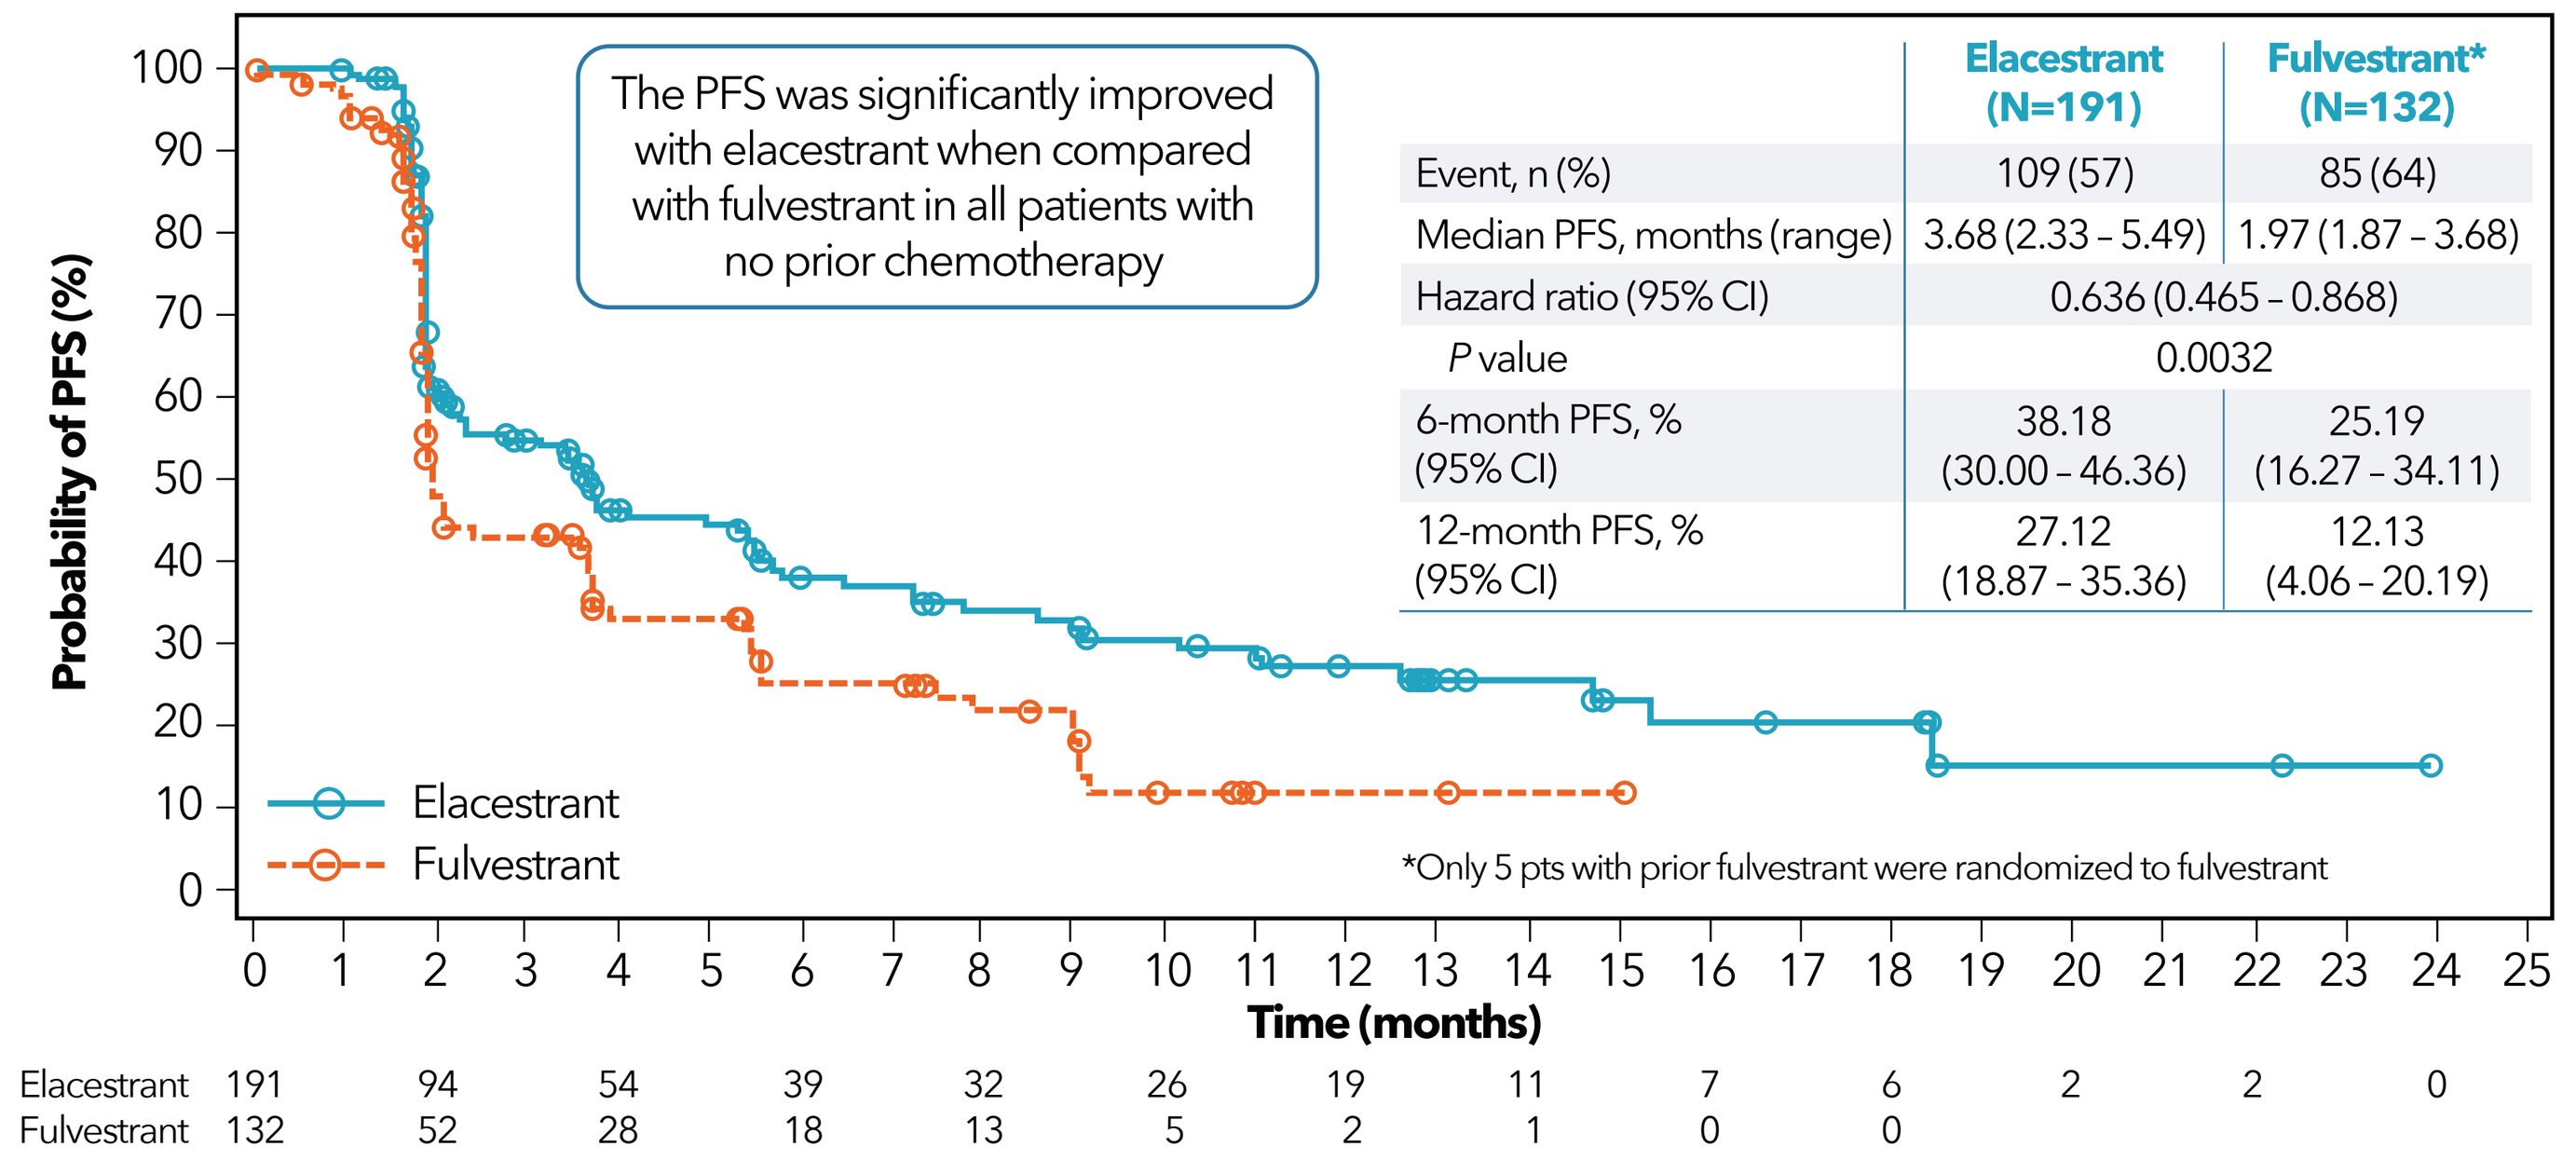

The PFS was significantly improved with elacestrant when compared with fulvestrant in all patients with no prior chemotherapy
Probability of PFS (%)
Time (months)
Elacestrant
Fulvestrant
Elacestrant 191 94 54 39 32 26 19 11 7 6 2 2 0
Fulvestrant 132 52 28 18 13 5 2 1 0 0

	Elacestrant (N=191)	Fulvestrant* (N=132)
Event, n (%)	109 (57)	85 (64)
Median PFS, months (range)	3.68 (2.33 – 5.49)	1.97 (1.87 – 3.68)
Hazard ratio (95% CI)	0.636 (0.465 – 0.868)	
P value	0.0032	
6-month PFS, % (95% CI)	38.18 (30.00 – 46.36)	25.19 (16.27 – 34.11)
12-month PFS, % (95% CI)	27.12 (18.87 – 35.36)	12.13 (4.06 – 20.19)

*Only 5 pts with prior fulvestrant were randomized to fulvestrant

PFS: elacestrant vs fulvestrant in patients with *mESR1* without prior chemotherapy (N=153)

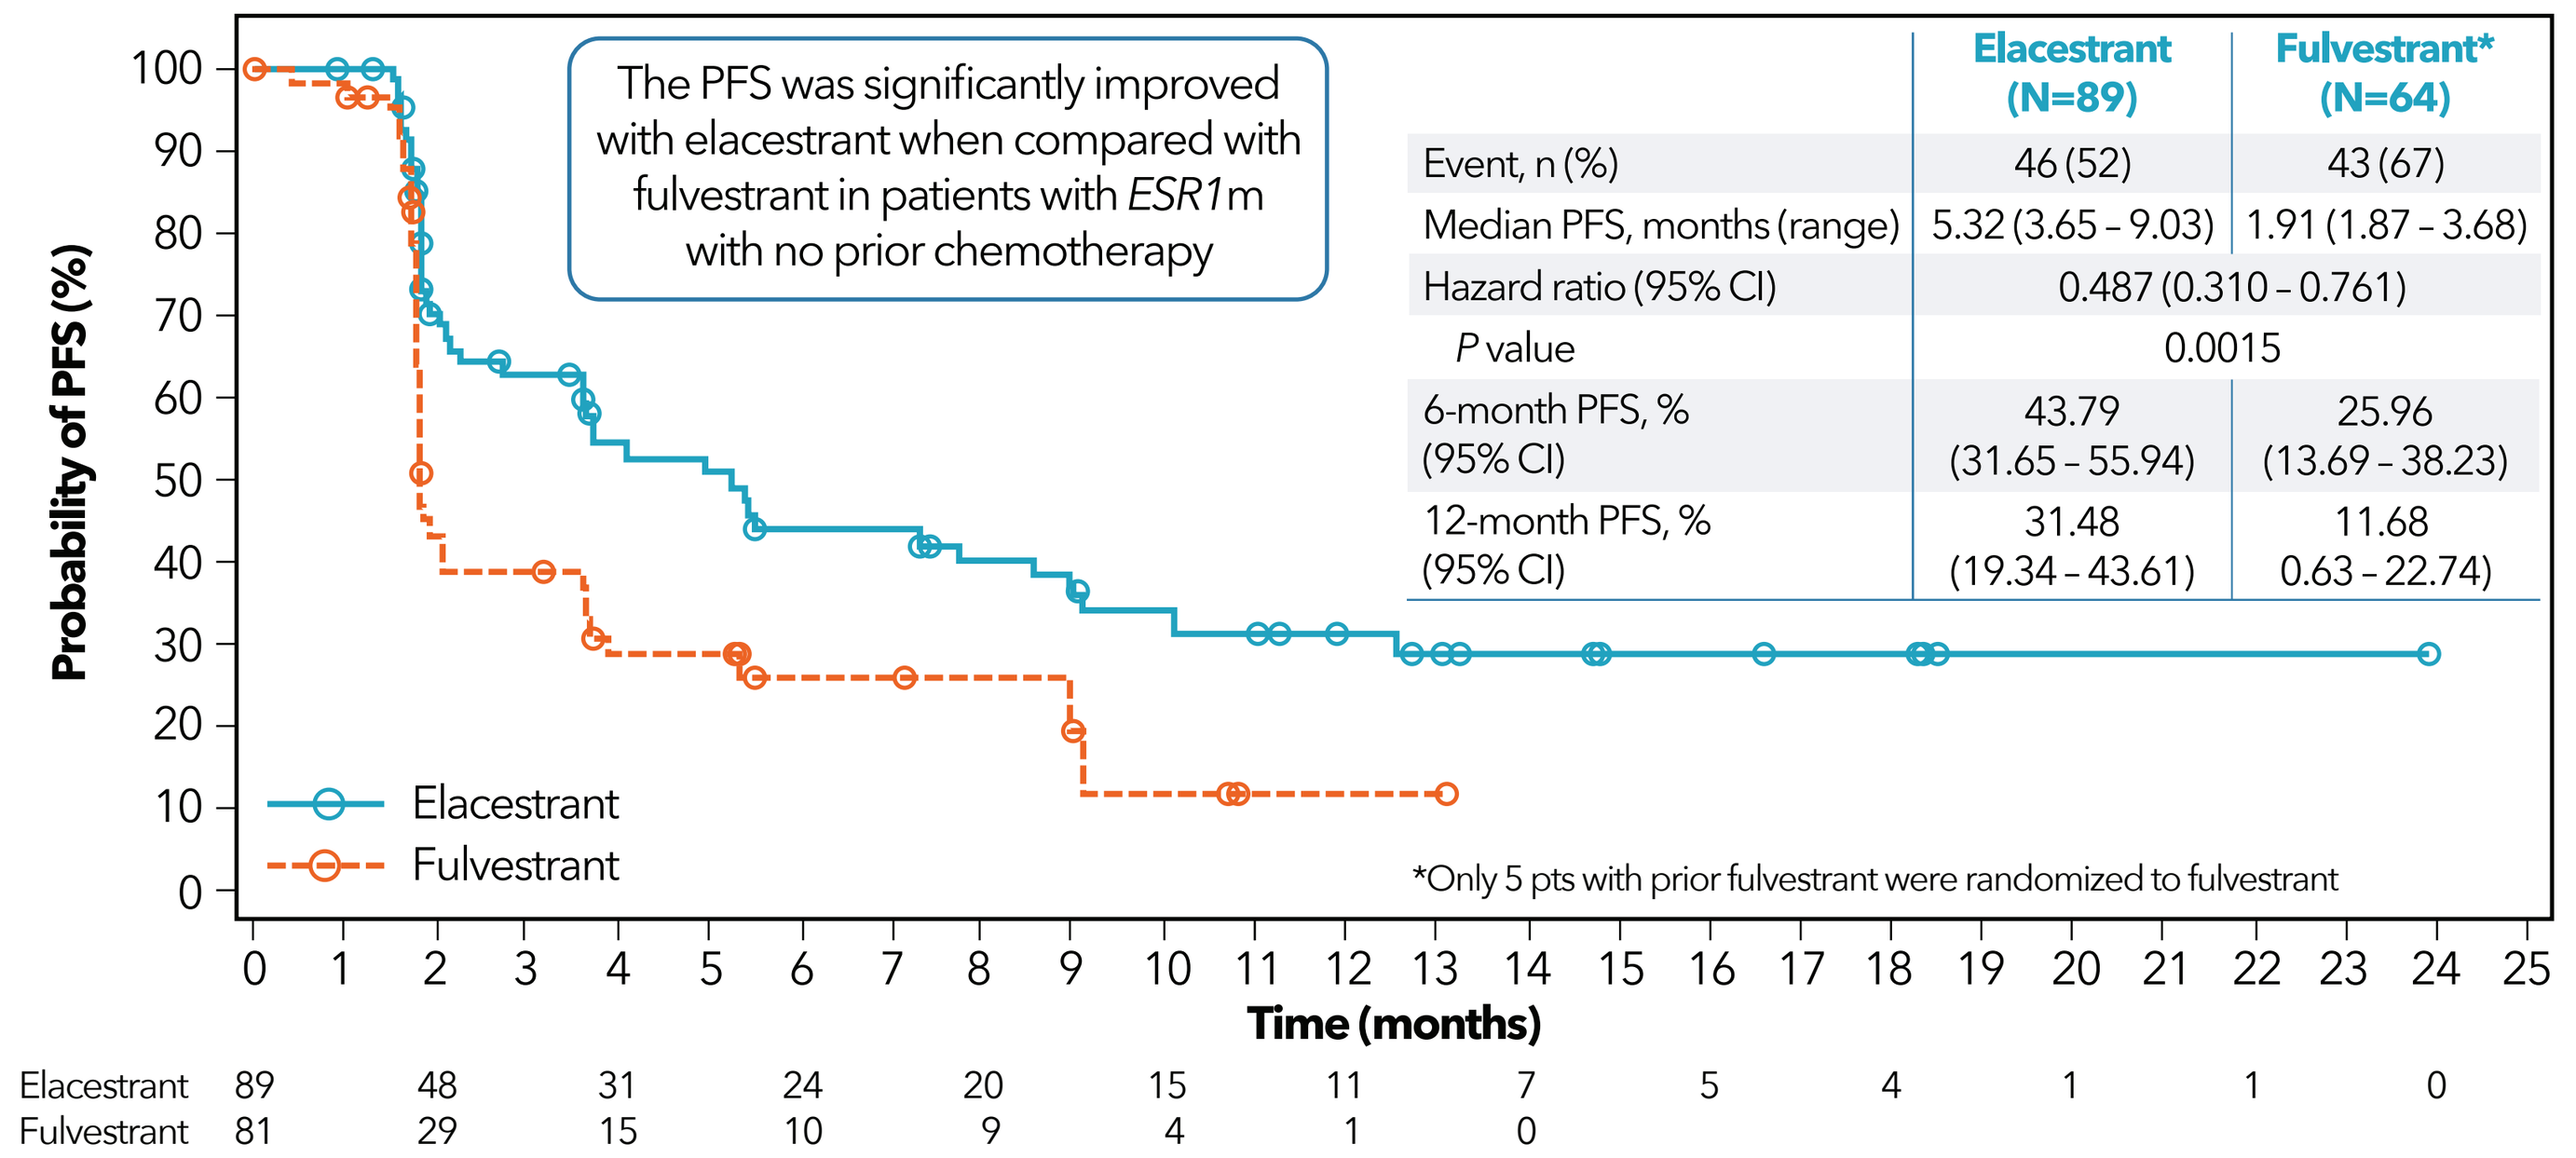

The PFS was significantly improved with elacestrant when compared with fulvestrant in patients with ESR1m with no prior chemotherapy
Probability of PFS (%)
Time (months)
Elacestrant
Fulvestrant
Elacestrant 89 48 31 24 20 15 11 7 5 4 1 1 0
Fulvestrant 81 29 15 10 9 4 1 0

	Elacestrant (N=89)	Fulvestrant* (N=64)
Event, n (%)	46 (52)	43 (67)
Median PFS, months (range)	5.32 (3.65 – 9.03)	1.91 (1.87 – 3.68)
Hazard ratio (95% CI)	0.487 (0.310 – 0.761)	
P value	0.0015	
6-month PFS, % (95% CI)	43.79 (31.65 – 55.94)	25.96 (13.69 – 38.23)
12-month PFS, % (95% CI)	31.48 (19.34 – 43.61)	11.68 0.63 – 22.74)

*Only 5 pts with prior fulvestrant were randomized to fulvestrant

Treatment-emergent adverse events (≥10% in either arm)

	Elacestrant N=189, n (%)		SOC Total N=175, n (%)		SOC Fulvestrant N=129, n (%)		SOC Aromatase inhibitor N=46, n (%)	
Preferred term	All Grades	Grade 3/4	All Grades	Grade 3/4	All Grades	Grade 3/4	All Grades	Grade 3/4
Nausea	64 (33.9%)	2 (1.1)	34 (19.4%)	–	21 (16.3%)	–	13 (28.3%)	–
Fatigue	36 (19.0%)	–	28 (16%)	–	21 (16.3%)	–	7 (15.2%)	–
Vomiting	33 (17.5%)	1 (0.5)	12 (6.9%)	–	9 (7.0%)	–	3 (6.5%)	–
Arthralgia	28 (14.8%)	–	30 (17.1%)	–	23 (17.8%)	–	7 (15.2%)	–
Decreased appetite	25 (13.2%)	1 (0.5)	13 (7.4%)	–	9 (7.0%)	–	4 (8.7%)	–
Back pain	25 (13.2%)	1 (0.5)	14 (8.0%)	–	10 (7.8%)	–	4 (8.7%)	–
Diarrhea	24 (12.7%)	–	19 (10.9%)	–	13 (10.1%)	–	6 (13.0%)	–
Headache	24 (12.7%)	1 (0.5)	21 (12%)	–	15 (11.6%)	–	6 (13.0%)	–
Hot flush	24 (12.7%)	–	15 (8.6%)	–	11 (8.5%)	–	4 (8.7%)	–
AST increased	23 (12.2%)	–	21 (12%)	–	16 (12.4%)	–	5 (10.9%)	–
Constipation	22 (11.6%)	–	11 (6.3%)	–	7 (5.4%)	–	4 (8.7%)	–
Dyspepsia	19 (10.1%)	–	5 (2.9%)	–	4 (3.1%)	–	1 (2.2%)	–

- Key treatment-related adverse events (AEs) in the no prior chemotherapy elacestrant group were nausea (25.9%), fatigue (12.7%), and hot flush (11.1%). There were no treatment-related deaths in either group.

CONCLUSIONS

- Among patients with ER+/HER2– mBC without prior chemotherapy, elacestrant significantly prolonged PFS compared to SOC endocrine therapy and showed favorable outcomes in this subgroup.
 - 31% reduction in the risk of progression or death with elacestrant vs SOC in all patients (HR=0.681 [95% CI: 0.520 – 0.891]; P=0.00388) and prolonged median PFS (3.68 vs 1.97 months).
 - 46% reduction in the risk of progression or death with elacestrant vs SOC in patients with *mESR1* (HR=0.535 [95% CI: 0.356 – 0.799]; P=0.00235) and prolonged median PFS (5.32 vs 1.91 months).
- In exploratory subgroup analyses, elacestrant significantly reduced the risk of progression or death and prolonged median PFS vs fulvestrant in all patients (HR=0.636 [95% CI: 0.465-0.868]; mPFS 3.68 vs 1.97 months), and in patients with *mESR1* (HR=0.487 (95% CI: 0.310-0.761; mPFS 5.32 vs 1.91 months).
- Elacestrant had a manageable safety profile consistent with other endocrine therapies.
- Final overall survival analysis of elacestrant vs SOC endocrine therapy expected late 2022/early 2023.
- Further elacestrant combinations in earlier lines and with other targeted therapies, including CDK4/6 and mTOR inhibitors, are ongoing/planned for patients with ER+/HER2- breast cancer.

REFERENCES:
1. National Comprehensive Cancer Network. NCCN clinical practice guidelines in oncology: breast cancer. Version 2.2022. https://www.nccn.org/professionals/physician_gls/pdf/breast.pdf. Updated December 20, 2021. Accessed March 24, 2022; **2.** Gennari A, et al. *Ann Oncol*. 2021;32:1475-1495; **3.** Burstein HJ, et al. *J Clin Oncol.* 2021;39:3959-3977; **4.** Jeselsohn R, et al. *Clin Cancer Res*. 2014;20:1757-1767; **5.** Moy B, et al. *J Clin Oncol*. 2021;39:3938-3958; **6.** Lindeman GJ, et al. *J Clin Oncol*. 2021;39 (suppl 15): abstr 1004; **7.** Turner NC, et al. *Lancet Oncol*. 2020;21:1296-1308; **8.** Di Leo A, et al. *Lancet Oncol*. 2018;19:87-100; **9.** André F, et al. *N Engl J Med*. 2019;38020:1929-1940;**10.** Bihani T, et al. *Clin Cancer Res*. 2017;23:4793-4804; **11.** Bidard FC, et al. *J Clin Oncol*. 2022: In press.

ACKNOWLEDGEMENTS:
We would like to thank the patients and their families/caregivers from 228 sites in 17 countries for participating in this trial. We would like to thank the investigators and their support staff who participated in this work. This study was sponsored by Radius Health, Inc. and co-funded by Menarini Group. Writing/Layout support provided by Phillips Gilmore Oncology Communications, Inc. funded by Menarini Group.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.
Contact Kaklamani@uthscsa.edu with questions.